ASGCT 28th Annual Meeting
Poster Number, 1397

An Improved Approach Enables Efficient and Cost-effective Directed Evolution of AAV Capsids in Non-human Primates via Different Delivery Routes

Gang Wang, Qingyu Deng, Jintao Wang, Tianyi Sun, Ruixue Chen, Sihan Wang, Wenbo Fu, Obulkasim Amar, Yangyang Zhong, Aliyam Ahtamu, Yan Jiao, Lei Wang

Byongen Therapeutics, Beijing, China Correspondence: gang.wang@byongen.com

HIGHLIGHTS

- **DynEvoLib enables multiplexed AAV screening in multiple tissues and delivery routes in single NHPs, accelerating capsid discovery.**
- **The muscle-tropic capsid BY087 demonstrates a 21-fold and 13-fold higher mRNA expression in skeletal and cardiac muscle, respectively, compared to AAV9.**
- **The CNS-tropic capsids BY213 and BY368 exhibit 30-fold and 98-fold higher mRNA expression in the brain, respectively, following intravenous and intrathecal administration.**

INTRODCTION

Directed evolution of AAV capsids in non-human primates (NHPs) is vital for clinical translation but is often limited by high costs and extensive animal use. To overcome these challenges, we developed DynEvoLib (Dynamic Evolution Library), a high-throughput platform that enables efficient, cost-effective, and multiplexed *in vivo* evolution of AAV capsids in NHPs via simultaneous delivery through multiple routes.

DynEvoLib employs barcoded libraries with degenerate peptide insertions, along with barcoded pools and peptide tags, to distinguish capsid variants. These libraries are co-delivered to a single animal through various routes, and mRNA enrichment using oligo-magnetic beads significantly enhances transcript recovery.

Using this platform, we identified AAV capsid variants with enhanced tissue tropism in rhesus macaques. Notably, the muscle-tropic BY087 demonstrated superior transduction in skeletal and cardiac muscles, while CNS-tropic variants BY213 and BY368 showed increased mRNA expression in the brain following intravenous and intrathecal delivery, respectively. These results highlight DynEvoLib's potential to generate tailored AAV vectors for targeted gene therapies.

METHODS

DynEvoLib-Based Capsid Directed Evolution via Multiple Delivery Routes

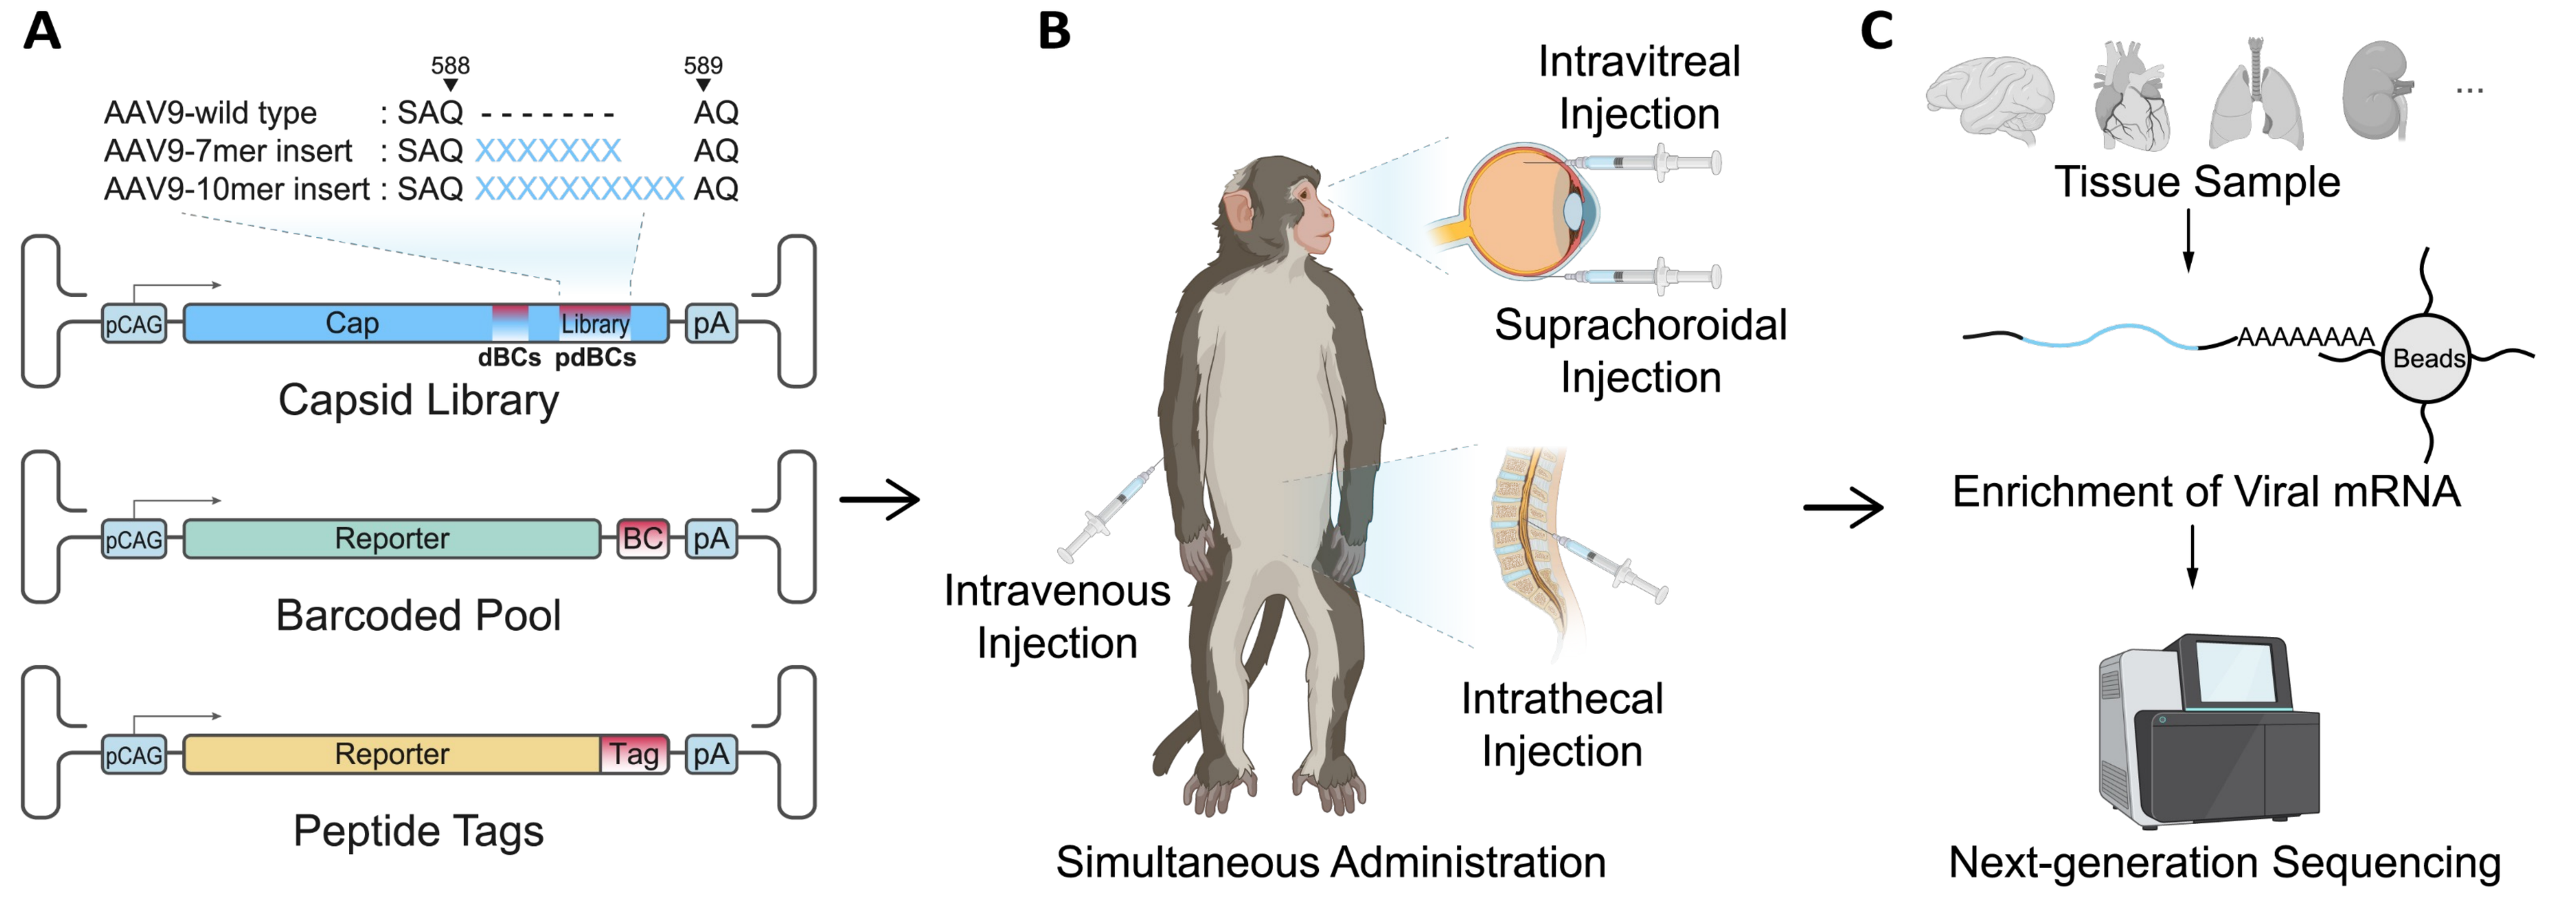
A
588
589
AAV9-wild type : SAQ - - - - - - - AQ
AAV9-7mer insert : SAQ XXXXXXX AQ
AAV9-10mer insert : SAQ XXXXXXXXXX AQ
pCAG
Cap
Library
pA
dBCs pdBCs
Capsid Library
Reporter
BC
Barcoded Pool
Tag
Peptide Tags
B
Intravitreal Injection
Suprachoroidal Injection
Intravenous Injection
Intrathecal Injection
Simultaneous Administration
C
Tissue Sample
AAAAAAAA
Beads
Enrichment of Viral mRNA
Next-generation Sequencing

(A) Three types of libraries or pools were used in DynEvoLib. First-round screening involved high-throughput capsid libraries with random or protein-derived peptides inserted into AAV9 capsids. Degenerate mutations flanking the insertion site created degenerate barcodes (dBCs) to distinguish libraries. Degenerate oligonucleotides generated peptide-derived degenerate barcodes (pdBCs) for subsequent rounds. mRNA expression of selected variants was quantified by deep sequencing of barcoded AAV pools. Finally, AAVs with multiple peptide tags were used to characterize variants at the DNA, RNA, and protein levels. **(B)** Libraries with different dBCs/pdBCs, barcoded pools, and peptide-tagged AAVs were administered to a single animal via multiple delivery routes. **(C)** Oligo-based magnetic beads enriched viral mRNA from total RNA, improving transcript acquisition. The mRNA enrichment strategy led to a 151-fold increase in viral transcript collection compared to regular single-tube RT-PCR.

RESULTS

Selection of Mouse Brain-Tropic Variants from Libraries, Barcoded Pools, and Peptide-Tagged AAVs

Up to 576 variants demonstrated brain transduction comparable to or exceeding that of the benchmark capsid AAV-F, with strong consistency across capsid libraries, barcoded pools, and peptide-tagged AAVs, including enhanced brain transduction and liver de-targeting.

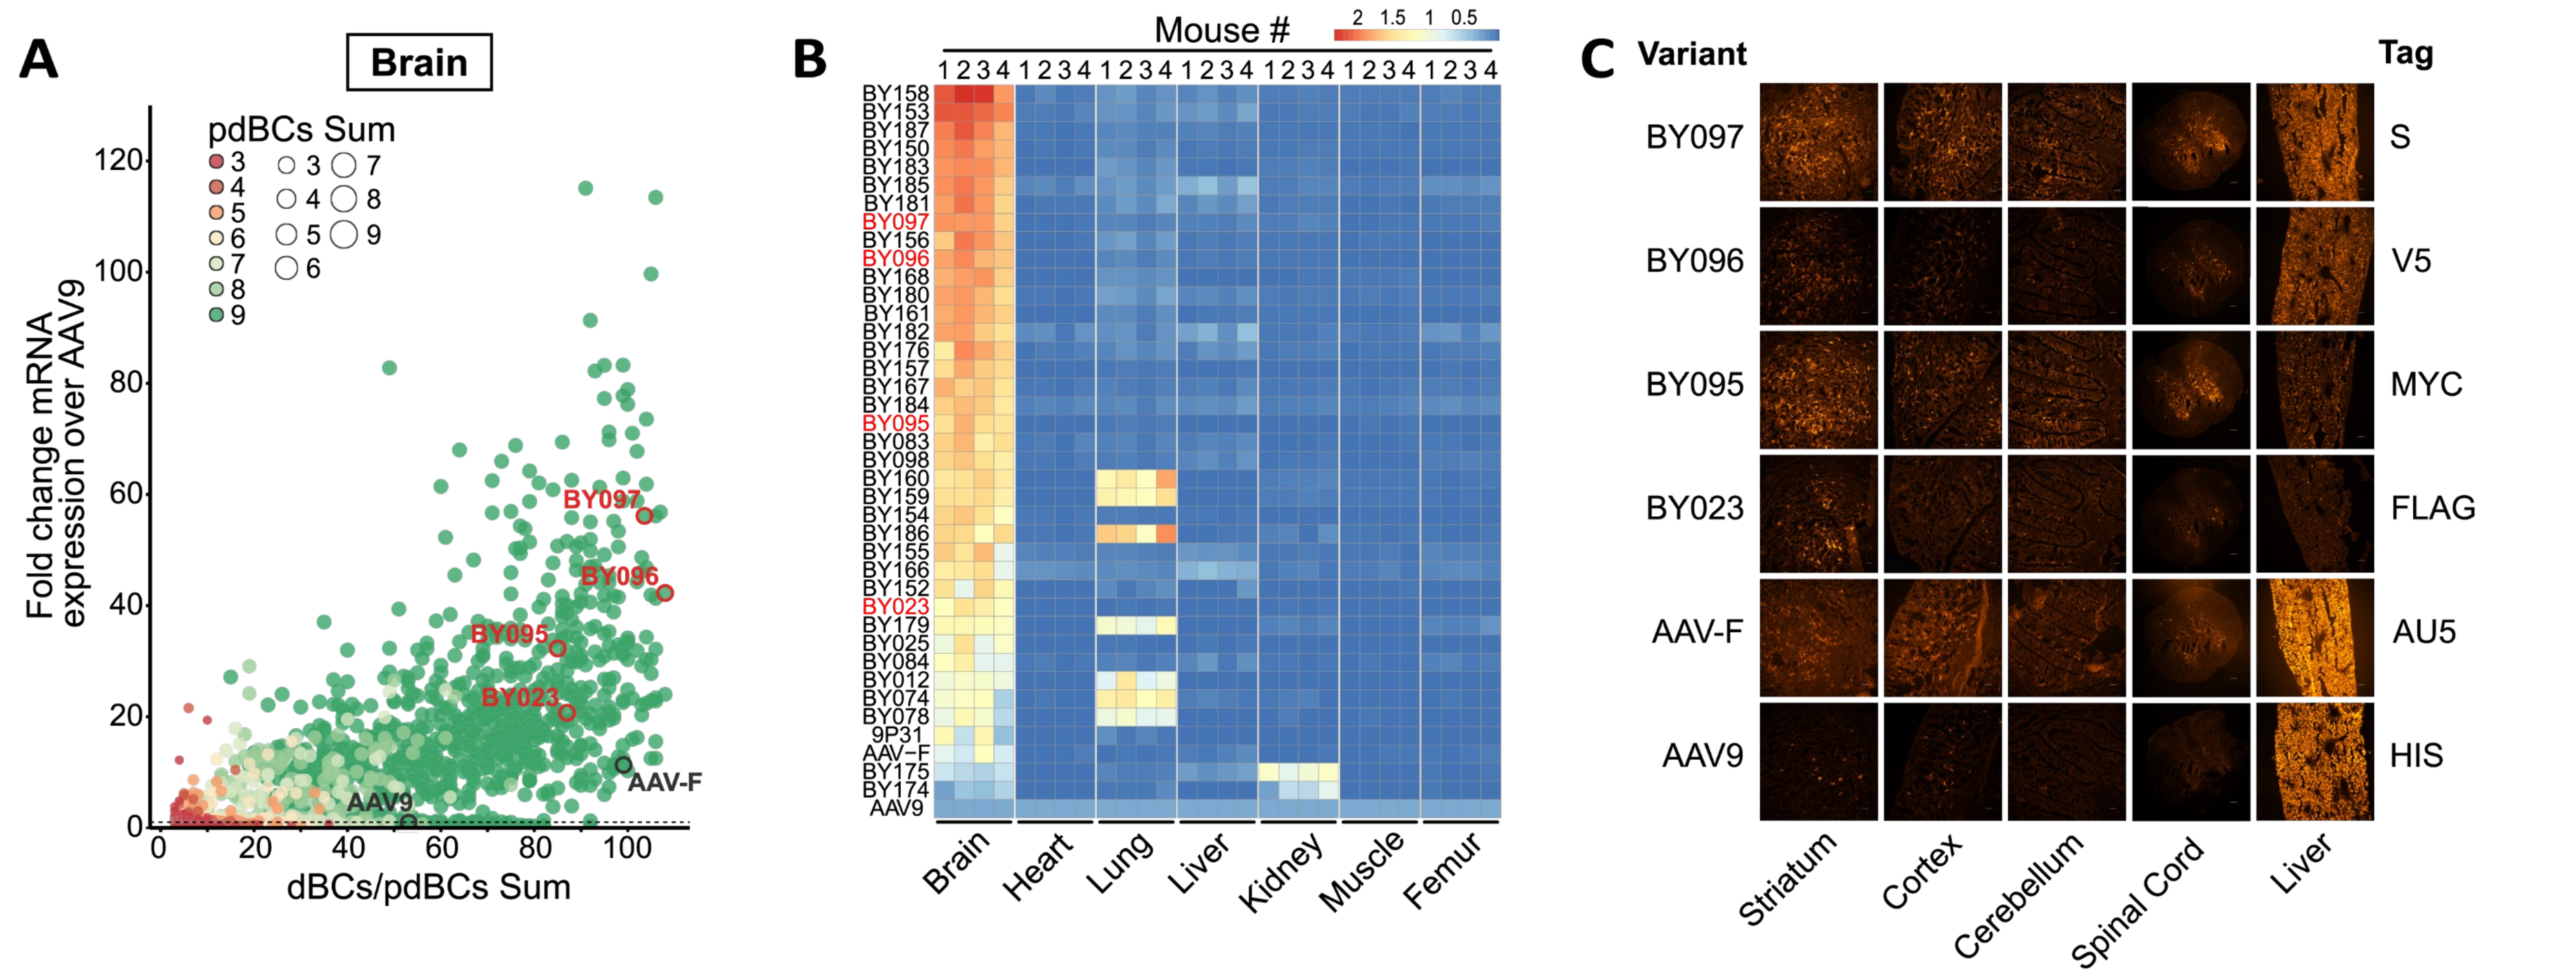
A
Brain
pdBCs Sum
Fold change mRNA expression over AAV9
dBCs/pdBCs Sum
BY097
BY096
BY095
BY023
AAV-F
AAV9
B
Mouse #
Brain
Heart
Lung
Liver
Kidney
Muscle
Femur
C
Variant
Tag
BY097
BY096
BY095
BY023
AAV-F
AAV9
S
V5
MYC
FLAG
AU5
HIS
Striatum
Cortex
Cerebellum
Spinal Cord
Liver

(A) Fold change in mRNA expression of variants over AAV9, total number (Sum) of dBCs/pdBCs combinations, and pdBCs in the brain of BALB/c mice (n = 3) 3 weeks after intravenous (IV) injection of the capsid library containing 30,182 variants selected from various tissues. **(B)** Heatmap showing Log_{10} fold changes in relative mRNA enrichment (normalized to AAV9 control) of variants in the brain and peripheral tissues of BALB/c mice (n = 4) 3 weeks after IV injection of the barcoded pool. **(C)** Peptide tags detected by immunofluorescence in cryosections of CNS and liver from BALB/c mice (n = 3) 3 weeks after IV injection of the peptide-tagged AAVs associated with CNS-tropic variants and AAV9, AAV-F controls. Scale bar, 100 µm.

Peptide-Tagged AAVs Screening Identifies Muscle-Tropic Variant BY087 in NHPs and Rodents

Several myotropic variants were identified in rhesus macaques via intravenous injection. BY087 increased skeletal and cardiac muscle mRNA expression by 21-fold and 13-fold, respectively, compared to AAV9, with corresponding protein increases, demonstrating strong muscle tropism in NHPs, mice, and rats.

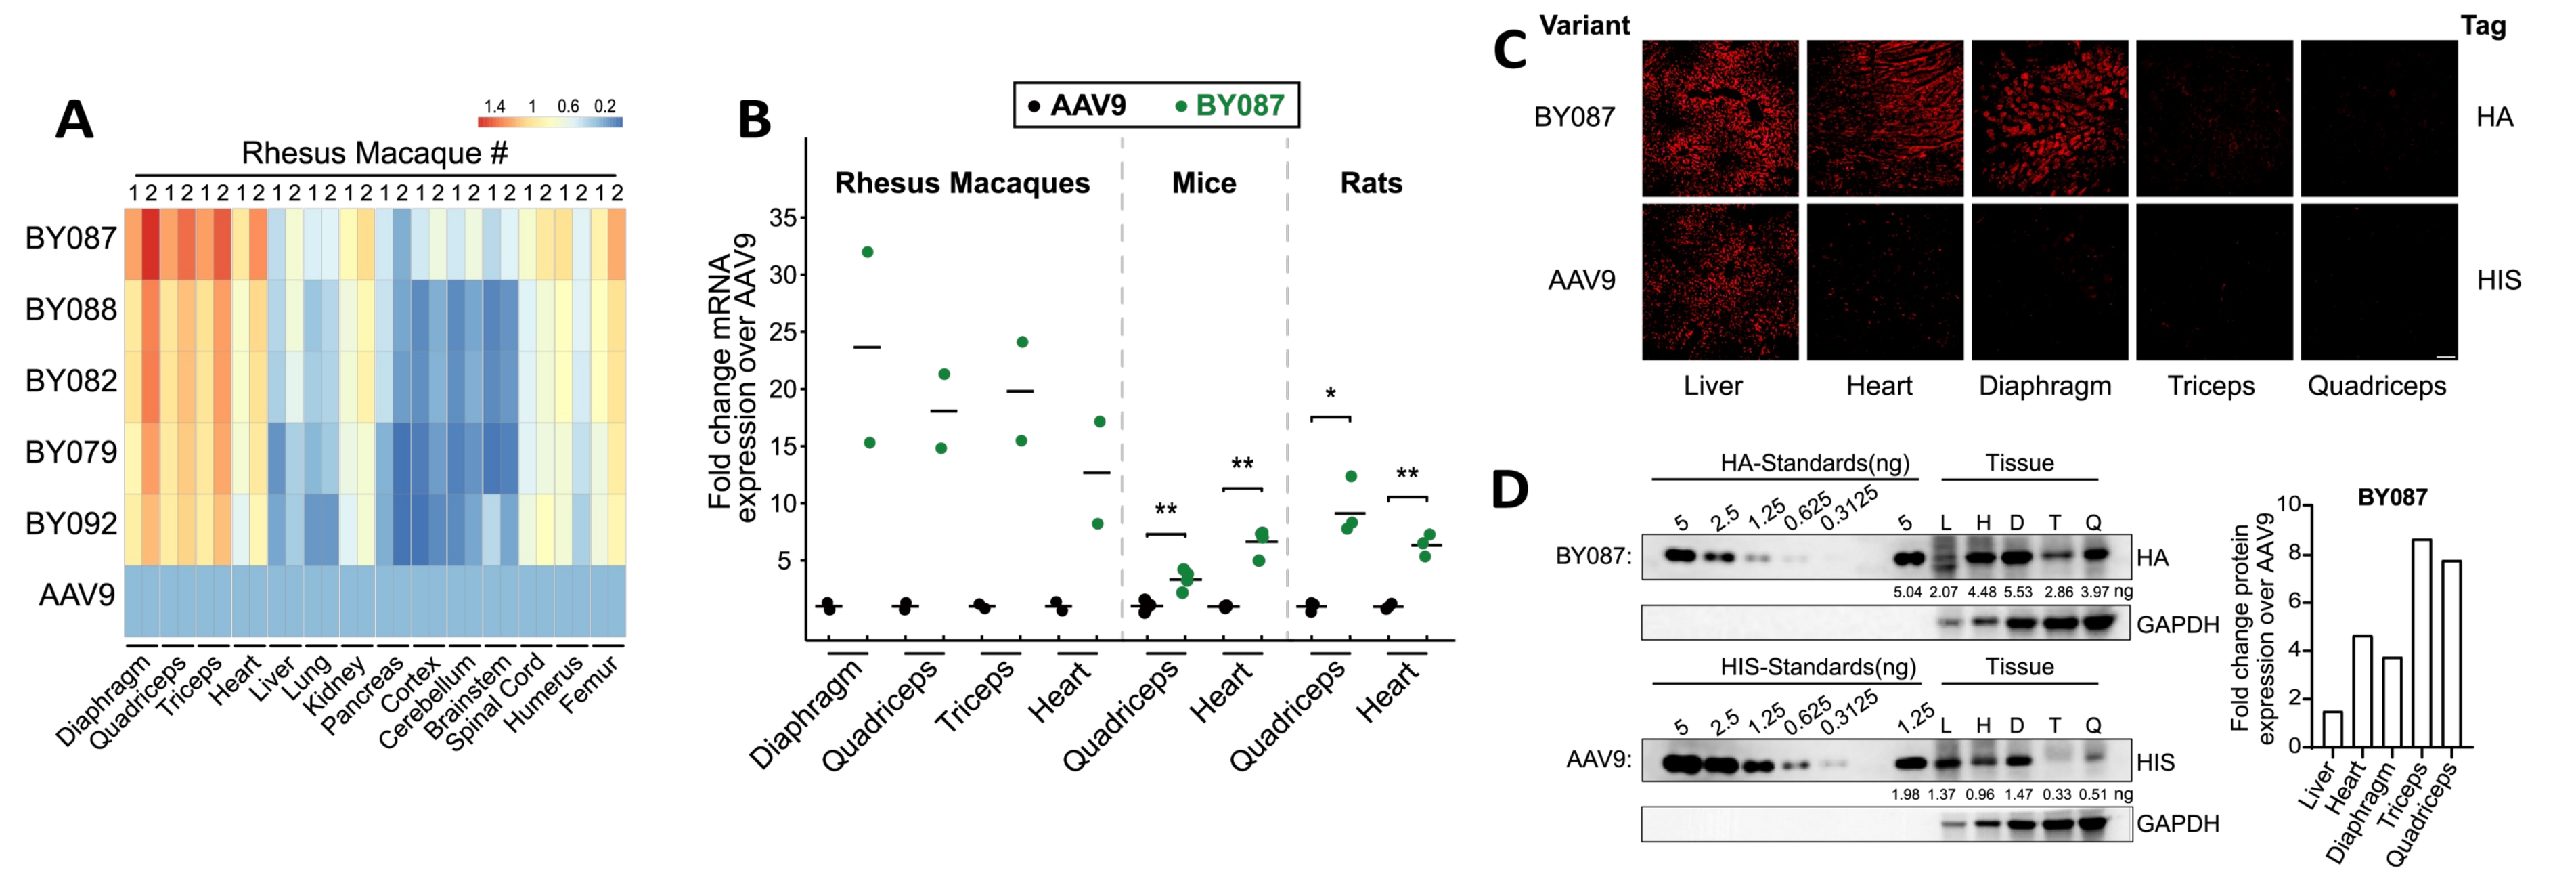
A
Rhesus Macaque #
BY087
BY088
BY082
BY079
BY092
AAV9
Diaphragm
Quadriceps
Triceps
Heart
Liver
Lung
Kidney
Pancreas
Cortex
Cerebellum
Brainstem
Spinal Cord
Humerus
Femur
B
AAV9
BY087
Rhesus Macaques
Mice
Rats
Fold change mRNA expression over AAV9
C
Variant
Tag
BY087
AAV9
HA
HIS
Liver
Heart
Diaphragm
Triceps
Quadriceps
D
HA-Standards(ng)
Tissue
BY087:
HA
GAPDH
HIS-Standards(ng)
AAV9:
HIS
Fold change protein expression over AAV9

(A) Heatmap showing Log_{10} fold changes in mRNA enrichment of myotropic AAV variants in skeletal muscle, heart, and other tissues of rhesus macaques (n = 2), 3 weeks after intravenous administration of peptide-tagged AAVs. Data are normalized to AAV9. **(B)** Fold change in BY087 mRNA expression relative to AAV9 in skeletal muscle and heart of rhesus macaques (n = 2), BALB/c mice (n = 4), and Sprague Dawley rats (n = 3), 3 weeks post-injection with the same peptide-tagged AAVs as in (A). Statistical significance: $^{**}p < 0.01$, $^{*}p < 0.05$. **(C)** Immunofluorescence detection of BY087 and AAV9 peptide tags in cryosections of liver, heart and in skeletal muscles from rhesus macaque #2. Scale bar, 200 µm. **(D)** Western blot analysis of BY087 and AAV9 peptide tags in skeletal muscles (D: diaphragm; Q: quadriceps; T: triceps), heart (H), and liver (L) from rhesus macaque #2. Protein expression levels were normalized to tag standards for quantification of total peptide-tagged protein and fold change analysis.

Intravenous Barcoded Pool Delivery Identifies CNS-Tropic Variants in NHPs

BY213 was identified as a top-performing variant, exhibiting an average 30-fold increase in mRNA expression across the CNS compared to AAV9, with performance comparable to or exceeding that of leading external engineered capsids.

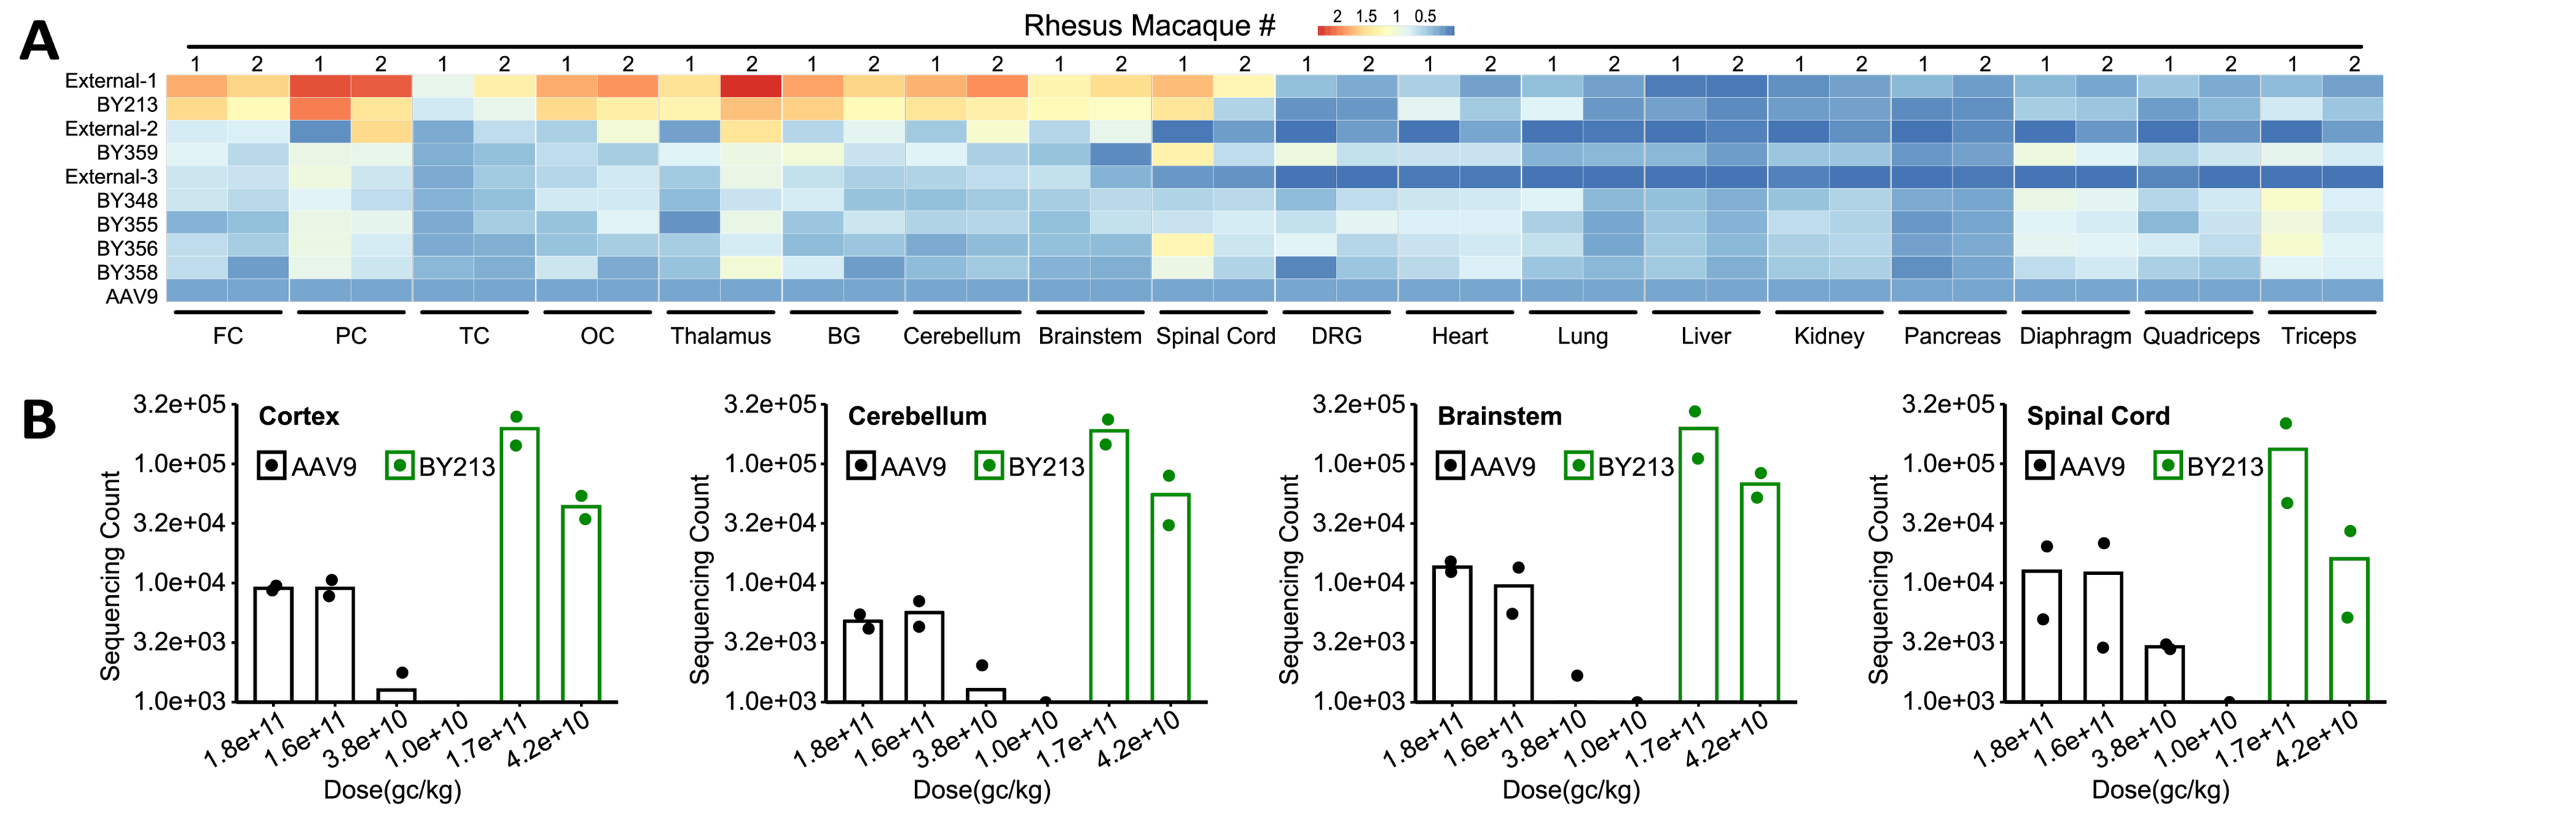
A
Rhesus Macaque #
External-1
BY213
External-2
BY359
External-3
BY348
BY355
BY356
BY358
AAV9
FC
PC
TC
OC
Thalamus
BG
Cerebellum
Brainstem
Spinal Cord
DRG
Heart
Lung
Liver
Kidney
Pancreas
Diaphragm
Quadriceps
Triceps
B
Cortex
Cerebellum
Brainstem
Spinal Cord
AAV9
BY213
Sequencing Count
Dose(gc/kg)

(A) Heatmap illustrating $\log_{10}$ fold changes in mRNA enrichment (normalized to AAV9 control) in the CNS of rhesus macaques (n = 2), three weeks after intravenous injection of the barcoded pool containing selected variants, AAV9, and external controls. **(B)** Normalized sequencing counts of BY213 mRNA expression relative to AAV9 across doses of barcoded AAVs in CNS regions. FC: frontal cortex, PC: parietal cortex, TC: temporal cortex, OC: occipital cortex, BG: basal ganglia, DRG: dorsal root ganglia.

Intrathecal Barcoded Pool Delivery Identifies CNS-Topic Variants in NHPs

Multiple variants were identified; BY368 demonstrated an average 98-fold increase in CNS-wide mRNA expression over AAV9—surpassing the benchmark AAV-DJ—and concurrently exhibited elevated off-target transduction in dorsal root ganglia.

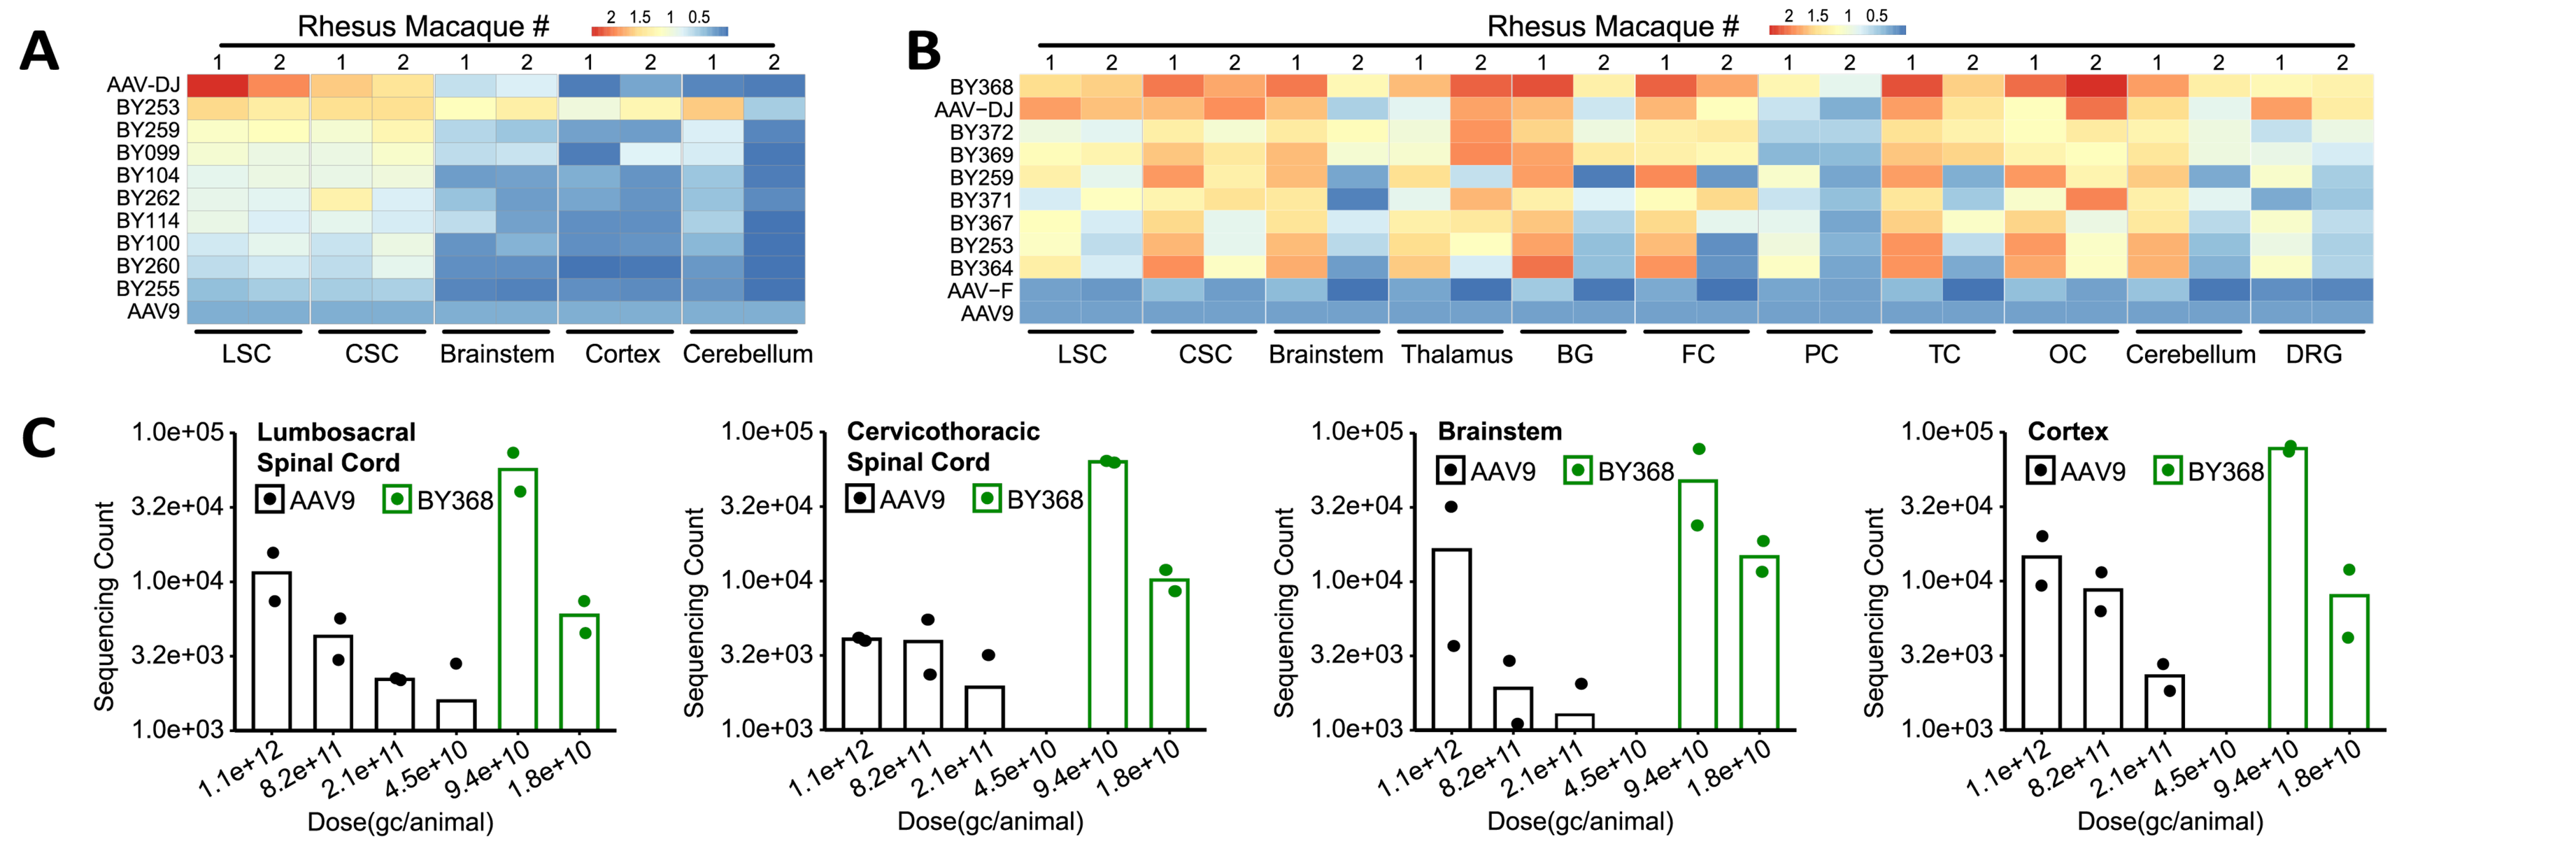
A
Rhesus Macaque #
AAV-DJ
BY253
BY259
BY099
BY104
BY262
BY114
BY100
BY260
BY255
AAV9
LSC
CSC
Brainstem
Cortex
Cerebellum
B
BY368
AAV-DJ
BY372
BY369
BY259
BY371
BY367
BY253
BY364
AAV-F
AAV9
Thalamus
BG
FC
PC
TC
OC
DRG
C
Lumbosacral Spinal Cord
Cervicothoracic Spinal Cord
Brainstem
Cortex
AAV9
BY368
Sequencing Count
Dose(gc/animal)

(A, B) Heatmaps showing $\log_{10}$ fold changes in relative mRNA enrichment (normalized to AAV9 control) in the CNS of rhesus macaques (n = 2), three weeks after intrathecal injection of the barcoded pool containing selected variants, alongside AAV9 and AAV-DJ controls (with AAV-F included in panel B only). Variants in panels (A) and (B) were derived from subsequent rounds of capsid library screening. **(C)** Normalized sequencing counts of BY368 mRNA expression relative to AAV9 across doses of barcoded AAVs in CNS regions. LSC: lumbosacral spinal cord, CSC: cervicothoracic spinal cord. Other abbreviations as in the previous figure.

CONCLUSION AND NEXT STEP

- DynEvoLib is a scalable NHP platform enabling multiplexed tissue/route screening in single subjects to accelerate cost-efficient AAV capsid discovery.
- The identification of BY087 (muscle-tropic) and BY213/BY368 (CNS-tropic via intravenous and intrathecal delivery) demonstrates the platform's ability to generate tailored AAV vectors.
- These capsid variants are undergoing further characterization, with BY087 currently being evaluated as a candidate for Duchenne Muscular Dystrophy gene therapy.